Commonly Prescribed Concomitant Medications and Clinical Safety Findings With Lenacapavir for Pre-Exposure Prophylaxis in PURPOSE 2

MeP16.1

PURPOSE 2

Anchalee Avihingsanon[1], Onyema Ogbuagu[2], Ian Frank[3], Cynthia Brinson[4], Gordon Crofoot[5], Javier Valencia[6], Christoph C Carter[7], Jenna Yager[7], Lillian B Brown[7], Pamela Wong[7], Jose Valdez Madruga[8], Pedro Cahn[9]

[1]HIV-NAT, Thai Red Cross AIDS and Infectious Disease Research Centre, and CE in Tuberculosis, Faculty of Medicine, Chulalongkorn University, Bangkok, Thailand; [2]Yale University, New Haven, CT, USA; [3]Perelman School of Medicine at the University of Pennsylvania, Philadelphia, PA, USA; [4]Central Texas Clinical Research, Austin, TX, USA; [5]Crofoot MD Clinic and Research Center, Houston, TX, USA; [6]Asociación Civil Impacta Salud y Educación, Sede San Miguel, Lima, Peru; [7]Gilead Sciences, Inc., Foster City, CA, USA; [8]Centro de Referência e Treinamento DST/AIDS-SP, São Paulo, Brazil; [9]Fundación Huésped, Buenos Aires, Argentina

Copies of this poster obtained through QR (Quick Response) are for personal use only and may not be reproduced without written permission of the authors

Conclusions

- There was no difference in rates of common adverse events (AEs) associated with phosphodiesterase type 5 (PDE5) inhibitors or statins between participants who did and did not use them together with lenacapavir (LEN) for pre-exposure prophylaxis (PrEP), even though LEN may increase concentrations of coadministered medications, such as PDE5 inhibitors and some statins, that are metabolized via cytochrome P450 3A (CYP3A)
 - — Rates of AEs associated with PDE5 inhibitor/statin use were low overall with the recommended dose adjustments per study protocol
- LEN, like other moderate CYP3A inhibitors, can be safely coadministered with PDE5 inhibitors and statins with appropriate monitoring and, in some cases, dose adjustment

Plain Language Summary

- Human immunodeficiency virus (HIV) prevention medication, also known as pre-exposure prophylaxis (or "PrEP"), helps to lower the chances of getting HIV
- Lenacapavir (LEN) is a long-acting type of PrEP that is given as an injection twice a year (every 6 months)
- In a clinical trial called PURPOSE 2, LEN worked very well to protect cisgender men, transgender women, transgender men, and gender nonbinary individuals who have sex with men from getting HIV
- LEN can slightly increase the amount of certain other medicines in the blood if they are taken together, including:
 - — Phosphodiesterase type 5 (PDE5) inhibitors (used for erectile dysfunction)
 - — Some statins (used to lower cholesterol levels)
- The higher blood levels of these other medicines can increase the chances of having side effects, like:
 - — Prolonged or painful erections, low blood pressure, fainting, or vision changes (with PDE5 inhibitors)
 - — Muscle pain, muscle damage, or changes in levels of liver enzymes (with statins)
- In the PURPOSE 2 study:
 - — Side effects that are commonly seen with PDE5 inhibitors or statins were rare
 - — None of the people taking PDE5 inhibitors with LEN had any of these side effects
 - — Among those taking statins with LEN, only one person reported muscle spasms and one reported changes in liver enzyme levels
- These results suggest that LEN can be safely used with PDE5 inhibitors or statins as long as healthcare providers regularly check and adjust the dosage if needed

Introduction

- PURPOSE 2 (NCT04925752) demonstrated superior efficacy of twice-yearly subcutaneous (SC) LEN versus daily oral emtricitabine/tenofovir disoproxil fumarate (F/TDF) as PrEP among cisgender men, transgender women, transgender men, and gender nonbinary individuals who have sex with partners assigned male at birth (Figure 1)[1]

Figure 1. Incidence of HIV Infections in PURPOSE 2[1]

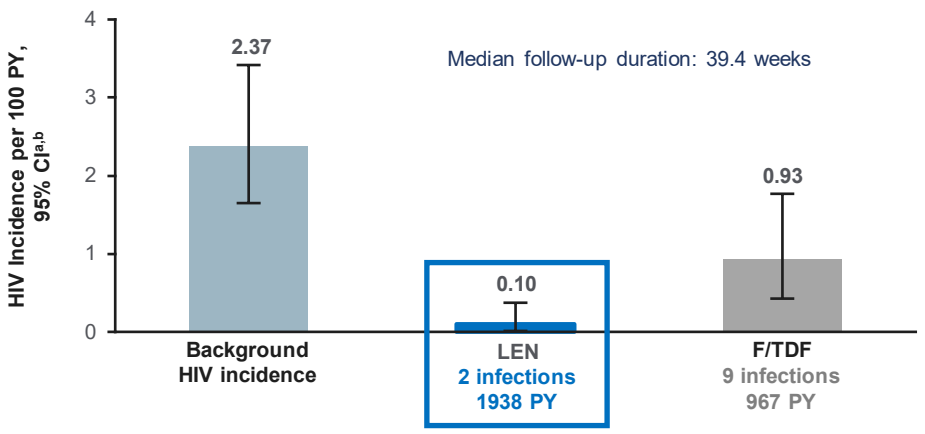
HIV Incidence per 100 PY, 95% CI[a,b]
4
3
2
1
0
2.37
Median follow-up duration: 39.4 weeks
0.10
0.93
Background HIV incidence
LEN
2 infections
1938 PY
F/TDF
9 infections
967 PY

[a]Overall n: background HIV incidence group, 4634; LEN, 2179; F/TDF, 1086.
[b]95% CIs: background HIV incidence group, 1.649-3.417; LEN, 0.012-0.373; F/TDF, 0.426-1.768.
CI, confidence interval; F/TDF, emtricitabine/tenofovir disoproxil fumarate; LEN, lenacapavir; PY, person-years.

- LEN is a moderate inhibitor of CYP3A (Figure 2)[2,3]
 - — Coadministered drugs that are metabolized by CYP3A (such as PDE5 inhibitors and statins) may therefore have slower clearance

Figure 2. LEN, a Moderate Inhibitor of CYP3A, May Increase Blood Levels of Coadministered Drugs That Are CYP3A Substrates[2,4]

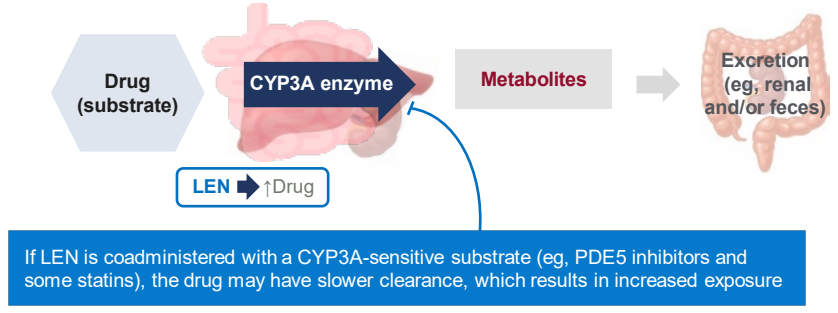
Drug (substrate)
CYP3A enzyme
Metabolites
Excretion (eg, renal and/or feces)
LEN ↑Drug
If LEN is coadministered with a CYP3A-sensitive substrate (eg, PDE5 inhibitors and some statins), the drug may have slower clearance, which results in increased exposure

CYP3A, cytochrome P450 3A; LEN, lenacapavir; PDE5, phosphodiesterase type 5.

- LEN may thus increase blood levels of coadministered medications that are metabolized by CYP3A, such as PDE5 inhibitors and some statins (Figure 3)[2]

Figure 3. PDE5 Inhibitors and Statins That Are Metabolized by CYP3A[2,5]

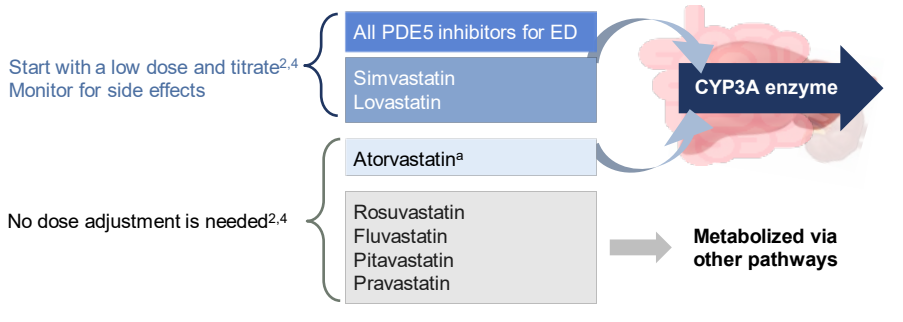
Start with a low dose and titrate[2,4]
Monitor for side effects
All PDE5 inhibitors for ED
Simvastatin
Lovastatin
CYP3A enzyme
Atorvastatin[a]
No dose adjustment is needed[2,4]
Rosuvastatin
Fluvastatin
Pitavastatin
Pravastatin
Metabolized via other pathways

[a]Although atorvastatin is metabolized by CYP3A, exposure is not impacted by coadministration with a moderate CYP3A inhibitor such as LEN because it is not a sensitive substrate of CYP3A.
CYP3A, cytochrome P450 3A; ED, erectile dysfunction; PDE5, phosphodiesterase type 5.

- Erectile dysfunction (ED) drugs such as PDE5 inhibitors may be used by people who could benefit from PrEP, including healthy young men without ED[6-9]
 - — Recreational use of PDE5 inhibitors has been reported by 22% of healthy young men[6] and 34% of gay and bisexual men[7]
 - — PDE5 inhibitor use has been reported in 12% to > 40% of PrEP users[8,9]

Objective

- To determine whether the rates of common AEs associated with PDE5 inhibitors or statins increased when these drugs were coadministered with LEN in PURPOSE 2

Methods

- PURPOSE 2 was a Phase 3, double-blind, randomized controlled trial in which participants were randomized 2:1 to receive twice-yearly SC LEN or daily oral F/TDF (Figure 4)
- Participants were seen at Weeks 4, 8, 13, and every 13 weeks thereafter; assessments for AEs occurred at each visit

Figure 4. PURPOSE 2 Study Design[1]

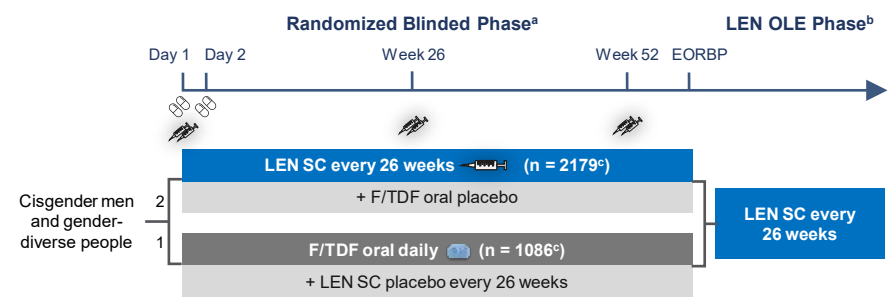
Randomized Blinded Phase[a]
LEN OLE Phase[b]
Day 1
Day 2
Week 26
Week 52
EORBP
Cisgender men and gender-diverse people
2
1
LEN SC every 26 weeks (n = 2179[c])
+ F/TDF oral placebo
F/TDF oral daily (n = 1086[c])
+ LEN SC placebo every 26 weeks
LEN SC every 26 weeks

Study population: Cisgender men and gender-diverse people ≥ 16 years old who are at risk of HIV acquisition[d]

Present analysis objectives

- To determine whether the rates of common AEs associated with PDE5 inhibitors or statins reported during the randomized blinded phase increased when these drugs were coadministered with LEN in PURPOSE 2

[a]Participants randomized to LEN received loading doses of two 300-mg tablets of LEN on each of Days 1 and 2 and SC LEN 927 mg on Day 1 and then every 26 weeks (± 7 days); participants randomized to F/TDF received matched placebos at these times.
[b]Participants randomized to LEN in the RBP who chose to participate in the LEN OLE Phase received SC LEN every 26 weeks (± 7 days) and had study visits every 13 weeks (± 7 days). Participants randomized to F/TDF in the RBP who chose to participate in the LEN OLE phase received SC LEN on LEN OLE Day 1 and every 26 weeks thereafter; these participants also received an oral LEN loading dose on LEN OLE Days 1 and 2 and had study visits at LEN OLE Day 1, Weeks 4 and 8 (± 2 days), Week 13 (± 7 days), and then every 13 weeks (± 7 days) thereafter.
[c]Included in the full analysis set for primary efficacy analyses.
[d]Cisgender men, transgender women, transgender men, and gender nonbinary individuals who have condomless receptive anal sex with partners assigned male at birth.
AE, adverse event; EORBP, end of randomized blinded phase; F/TDF, emtricitabine/tenofovir disoproxil fumarate; LEN, lenacapavir; OLE, open-label extension; PDE5, phosphodiesterase type 5; RBP, randomized blinded phase; SC, subcutaneous.

- Dose adjustments and/or monitoring of concomitant medications with potential drug-drug interactions with LEN were recommended during the study (Table 1)
- The frequency of AEs commonly associated with increased statin or PDE5 inhibitor exposure was assessed by study drug and baseline PDE5 inhibitor/statin use in the overall study population
 - — PDE5 inhibitor-associated AEs: Priapism, painful erection, hypotension, syncope, vision changes, and hearing loss
 - — Statin-associated AEs: Myalgia, rhabdomyolysis, muscle pain or discomfort, increased creatine phosphokinase levels, and increased liver function test values

Table 1. PURPOSE 2 Protocol Guidance on Coadministration of PDE5 Inhibitors and Statins With LEN

Coadministered Drug	Interaction Potential With LEN	Dosing Recommendations
PDE5 inhibitors	Concentrations of PDE5 inhibitors may increase with LEN	• Maximum permitted doses: – Sildenafil ≤ 25 mg in 48 hours – Vardenafil ≤ 2.5 mg in 72 hours – Tadalafil ≤ 10 mg in 72 hours
Statins	Concentrations of some statins may increase with LEN • Careful monitoring of signs or symptoms of muscle weakness or myopathy is recommended	• Maximum permitted doses: – Simvastatin 10 mg – Lovastatin 20 mg – Atorvastatin[a] 40 mg

[a]This maximum dose was recommended in the study out of caution for a potential drug-drug interaction, but an *a priori* dose adjustment of atorvastatin is not needed with LEN because it is a moderate, and not a sensitive, CYP3A substrate.
CYP3A, cytochrome P450 3A; LEN, lenacapavir; PDE5, phosphodiesterase type 5.

Results

- At baseline:
 - — PDE5 inhibitor use was reported by 29/2183 (1.3%) participants receiving LEN and 16/1088 (1.5%) receiving F/TDF
 - — Statin use was reported by 40/2183 (1.8%) participants receiving LEN and 24/1088 (2.2%) receiving F/TDF
- Frequency of common AEs associated with PDE5 inhibitors and statins are summarized in Tables 2 and 3
- No PDE5 inhibitor-associated AEs were reported among participants receiving PDE5 inhibitors at baseline
- Among participants receiving statins at baseline:
 - — Muscle spasms were reported by 1 (2.5%) participant in the LEN arm; muscle spasms and pain in extremity were each reported by 1 (4.2%) participant in the F/TDF arm
 - — Increased liver enzymes were reported by 1 (2.5%) participant in the LEN arm and none in the F/TDF arm
 - — Myalgia, rhabdomyolysis, and increased creatine phosphokinase levels were not reported

Table 2. No Increase in Common AEs Associated With PDE5 Inhibitors in PURPOSE 2

	All Participants (N = 3271)			
	Baseline PDE5 Inhibitor Use			
	Yes (n = 45)		No (n = 3226)	
	LEN (n = 29)	F/TDF (n = 16)	LEN (n = 2154)	F/TDF (n = 1072)
Priapism	0	0	0	1 (< 0.1)
Painful erection	0	0	0	1 (< 0.1)
Hypotension	0	0	2 (< 0.1)	2 (0.2)
Syncope	0	0	17 (0.8)	14 (1.3)
Vision change[a]	0	0	4 (0.2)	0
Hearing loss[b]	0	0	1 (< 0.1)	0

Data are n (%). AEs are based on AE Preferred Terms coded using MedDRA, Version 27.0.
[a]Includes vision blurred, visual acuity reduced, and vision impairment.
[b]Hearing loss based on the AE high-level term.
AE, adverse event; LEN, lenacapavir; MedDRA, Medical Dictionary for Regulatory Activities; PDE5, phosphodiesterase type 5; F/TDF, emtricitabine/tenofovir disoproxil fumarate.

Table 3. No Increase in Common AEs Associated With Statins in PURPOSE 2

	All Participants (N = 3271)			
	Baseline Statin Use			
	Yes (n = 64)		No (n = 3207)	
	LEN[a] (n = 40)	F/TDF[b] (n = 24)	LEN (n = 2143)	F/TDF (n = 1064)
Myalgia	0	0	32 (1.5)	13 (1.2)
Rhabdomyolysis	0	0	4 (0.2)	1 (< 0.1)
Muscle/extremity pain or discomfort[c]	1 (2.5)	2 (8.3)	12 (0.6)	7 (0.7)
Increased CPK	0	0	7 (0.3)	5 (0.5)
Increased liver enzymes[d]	1 (2.5)	0	14 (0.7)	8 (0.8)

Data are n (%). AEs are based on AE Preferred Terms coded using MedDRA, Version 27.0.
[a]Atorvastatin (n = 17), rosuvastatin (n = 15), simvastatin (n = 10). Number of summed participants differs from total if participants switched treatments.
[b]Atorvastatin (n = 14), rosuvastatin (n = 6), simvastatin (n = 5). Number of summed participants differs from total if participants switched treatments.
[c]Includes pain in extremity, musculoskeletal pain, musculoskeletal discomfort, and muscle spasms.
[d]Increases reported as AEs, not based on reported laboratory tests; includes hypertransaminasemia, increased ALT, increased AST, increased hepatic enzymes, abnormal transaminases, increased bilirubin, increased unconjugated bilirubin, and hyperbilirubinemia.
AE, adverse event; ALT, alanine aminotransferase; AST, aspartate aminotransferase; CPK, creatine phosphokinase; LEN, lenacapavir; MedDRA, Medical Dictionary for Regulatory Activities; F/TDF, emtricitabine/tenofovir disoproxil fumarate.

Study Limitations

- The numbers of participants reporting concomitant PDE5 inhibitor or statin use were low, as were the rates of common AEs associated with statin/PDE5 inhibitor use
- Any baseline statin use was included in this analysis, regardless of whether the statin was a CYP3A substrate
- AE causality cannot be determined from this dataset and AEs may be attributable to causes other than concomitant medication use

References: 1. Kelley CF, et al. *N Engl J Med*. 2025;392:1261-76. **2**. Gilead Sciences. https://www.gilead.com/-/media/files/pdfs/medicines/hiv/yeztugo/yeztugo_pi.pdf (accessed Aug. 13, 2025). **3**. Begley R, et al. Oral 89 presented at: CROI; March 6-10, 2021; Virtual. **4**. Bekker LG et al. Session OAC05 presented at: the 13th IAS Conference on HIV Science; July 13-17, 2025; Kigali, Rwanda. **5**. University of Liverpool. https://www.hiv-druginteractions.org/checker (accessed Aug. 13, 2025). **6**. Bechara A, et al. *J Sex Med*. 2010;7:3736-42. **7**. Hammoud MA, et al. *J Sex Med*. 2017;14(suppl. 4):e265. **8**. De La Mora L, et al. *AIDS Behav*. 2022;26:4055-62. **9**. Uhrmacher M, et al. *Front Public Health*. 2022;10:930208.

Acknowledgments: We extend our thanks to the PURPOSE 2 participants, their families, and all participating investigators. This study was funded by Gilead Sciences, Inc. Medical writing support was provided by Samantha Santangelo, PhD (Aspire Scientific Ltd, UK), and was funded by Gilead Sciences, Inc.

Disclosures: AA reports research grants to her institute from Gilead Sciences, Inc., MSD, ViiV Healthcare/GSK, and Roche; travel support from Gilead Sciences, Inc.; and unpaid roles with the Thai AIDS Society, WHO SEARO Advisory Group, and Thailand National Committees on ART, Hepatitis, and TB/TPT. **OO** reports receipt of an NIAID research grant, serving on advisory boards for Gilead Sciences, Inc. and ViiV Healthcare, and being an unpaid member of the Department of Health and Human Services' Panel for HIV Treatment Guidelines for Adults and Adolescents. **IF** reports research support from Gilead Sciences, Inc. and a research grant to his institution (University of Pennsylvania); grants or contracts from Janssen, Moderna, the National Institutes of Health, Pfizer, Sanofi, and Vir Biotechnology; and serving on a data and safety monitoring board for Johnson & Johnson and an advisory board for ViiV Healthcare. **CB** has no other disclosures to report. **GC** has no other disclosures to report. **JV** has no other disclosures to report. **CCC**, **JY**, **LBB**, and **PW** are employees of and hold shares in Gilead Sciences, Inc. **JVM** reports advisory board participation and speaker support from Gilead Sciences, Inc., ViiV Healthcare/GSK, and Janssen, and travel support from Janssen. **PC** reports advisory boards for Gilead Sciences, Inc. and ViiV Healthcare.

Correspondence: Anchalee Avihingsanon, anchaleea2009@gmail.com.

20th European AIDS Conference (EACS); October 15–18, 2025; Paris, France